Ex Vivo Treatment By Mitapivat, an Allosteric Pyruvate Kinase Activator, Reduced Hemolysis and Reactive Oxygen Species in Red Blood Cells of Non-Transfusion Dependent Hemolytic Anemic Patients Due to β-Thalassemia/Hb E Disease

T. SUKSANGPLENG[1], K. TACHAVANICH[2] and V. VIPRAKASIT[2]

1. Siriraj Thalassemia Center, Faculty of Medicine Siriraj Hospital, Mahidol University, Bangkok, Thailand
2. Department of Pediatrics, Faculty of Medicine Siriraj Hospital, Mahidol University, Bangkok, Thailand

INTRODUCTION

The β-thalassemia/Hemoglobin E (β-thal/Hb E) disease, an inherited disorder that combines the β-globin gene mutations, including β^o and β^+, and the Hb E variant, is frequently found in Southeast Asian populations, including Thailand. Pathophysiology of red blood cells (RBCs) of anemic patients with β-thal/Hb E is caused by a reduction of β-globin synthesis from β^o or β^+mutations leading to an unbalanced excess of α-globin chains. Free α-globin chains trigger a cascade of events through generation of intracellular reactive oxygen species (ROS) leading to peripheral RBC hemolysis, resulting in the moderate severity of β-thal/Hb E. Mitapivat (AG-348), an allosteric activator of pyruvate kinase (PK), is being evaluated for its efficacy and safety as a potential therapy for the treatment of thalassemia (NCT04770753 and NCT04770779). We are currently studying the effects of mitapivat on RBC pathophysiology and production of adenosine triphosphate (ATP), the energy carrier molecule derived from the glycolytic pathway by PK activity, in the RBCs of β-thal/Hb E patients.

AIM

To evaluate the effects of mitapivat on RBC pathophysiology, including intracellular ROS, hemolysis, and ATP production, in RBCs of non-transfusion dependent hemolytic anemia patients with β-thal/Hb E disease (NTDT) in an *ex vivo* setting.

METHOD

A 2-mL EDTA whole blood sample was collected from non-regularly transfused patients who were diagnosed with β-thal/Hb E disease by multiplex PCR methods to specifically detect 9 common β-globin gene mutations found in the Thai population. RBCs were separated by an α-cellulose bead column and prepared into a suspension of 1 x 10^7 RBCs/mL in a complete culture medium. RBC suspension was incubated with mitapivat (Agios Pharmaceuticals, Inc.) at 37ºC for 24 h. The mitapivat-treated RBCs were harvested to measure ATP levels. Oxidative stress was evaluated by measuring the mean fluorescent intensity (MFI) of intracellular ROS. The culture medium was collected to measure the Hb released, which represented RBC hemolysis.

RESULTS

The 25 non-regularly transfused patients with β-thal/Hb E disease, including 20 and 5 cases with $\beta^o\beta^E$ and $\beta^+\beta^E$ genotypes, respectively, had chronic anemia with the mean (± standard deviation; SD) Hb concentration of 8.0±0.6 and 8.9±1.0 gm/dL, respectively **(Table 1)**. After treatment with various concentrations between 0.002 to 10 µM mitapivat, the ATP level detected in the RBCs of patients with $\beta^o\beta^E$ and $\beta^+\beta^E$ genotypes significantly increased in a concentration-dependent manner from 100% to 120% and from 100% to 150%, respectively, compared to the baseline ATP level of 0.1% DMSO solvent control **(Figure 1A)**. The MFI of ROS in mitapivat-treated $\beta^o\beta^E$ and $\beta^+\beta^E$ RBCs decreased from 90% to 70% compared to baseline MFI of solvent control, indicating a reduction in oxidative stress **(Figure 1B)**. Moreover, reduced intracellular ROS found in mitapivat-treated RBCs corresponded to reduced hemolysis as reflected by Hb released from treated RBCs in the culture medium. A decrease in the amount of Hb released (µg/mL) in a concentration-dependent manner was found from 90% to 70% compared to the baseline Hb released of solvent control **(Figure 1C)**.

CONCLUSIONS

This *ex vivo* study showed that mitapivat could reduce hemolysis and decrease intracellular ROS in RBCs of non-transfusion dependent β-thal/Hb E disease patients presented with moderately to severe hemolytic anemia. Moreover, mitapivat promoted ATP production, which supported energy for RBC survival in the blood of β-thal/Hb E patients. Data obtained from this study supports the current clinical trials of mitapivat in thalassemia to ameliorate clinical anemia in patients due to β-globin gene mutations by improving RBC physiology and decreasing peripheral hemolysis.

REFERENCES

Kung C, et al. AG-348 enhances pyruvate kinase activity in red blood cells from patients with pyruvate kinase deficiency. *Blood 2017; 130: 1347-56.*

van Beers EJ, et al. Mitapivat, a novel pyruvate kinase activator, for the treatment of hereditary hemolytic anemias. *Ther Adv Hematol 2021; 12: 1–11.*

Matte A, et al. The pyruvate kinase activator mitapivat reduces hemolysis and improves anemia in a β-thalassemia mouse model. *J Clin Invest 2021; 131: e144206.*

Figure 1 Effects of *ex vivo* mitapivat treatment on ATP production (A), intracellular ROS (B), and hemolysis (C) in treated RBCs of 5 NTDT patients with $\beta^o\beta^E$ genotypes and 2 NTDT patients with $\beta^+\beta^E$ genotypes

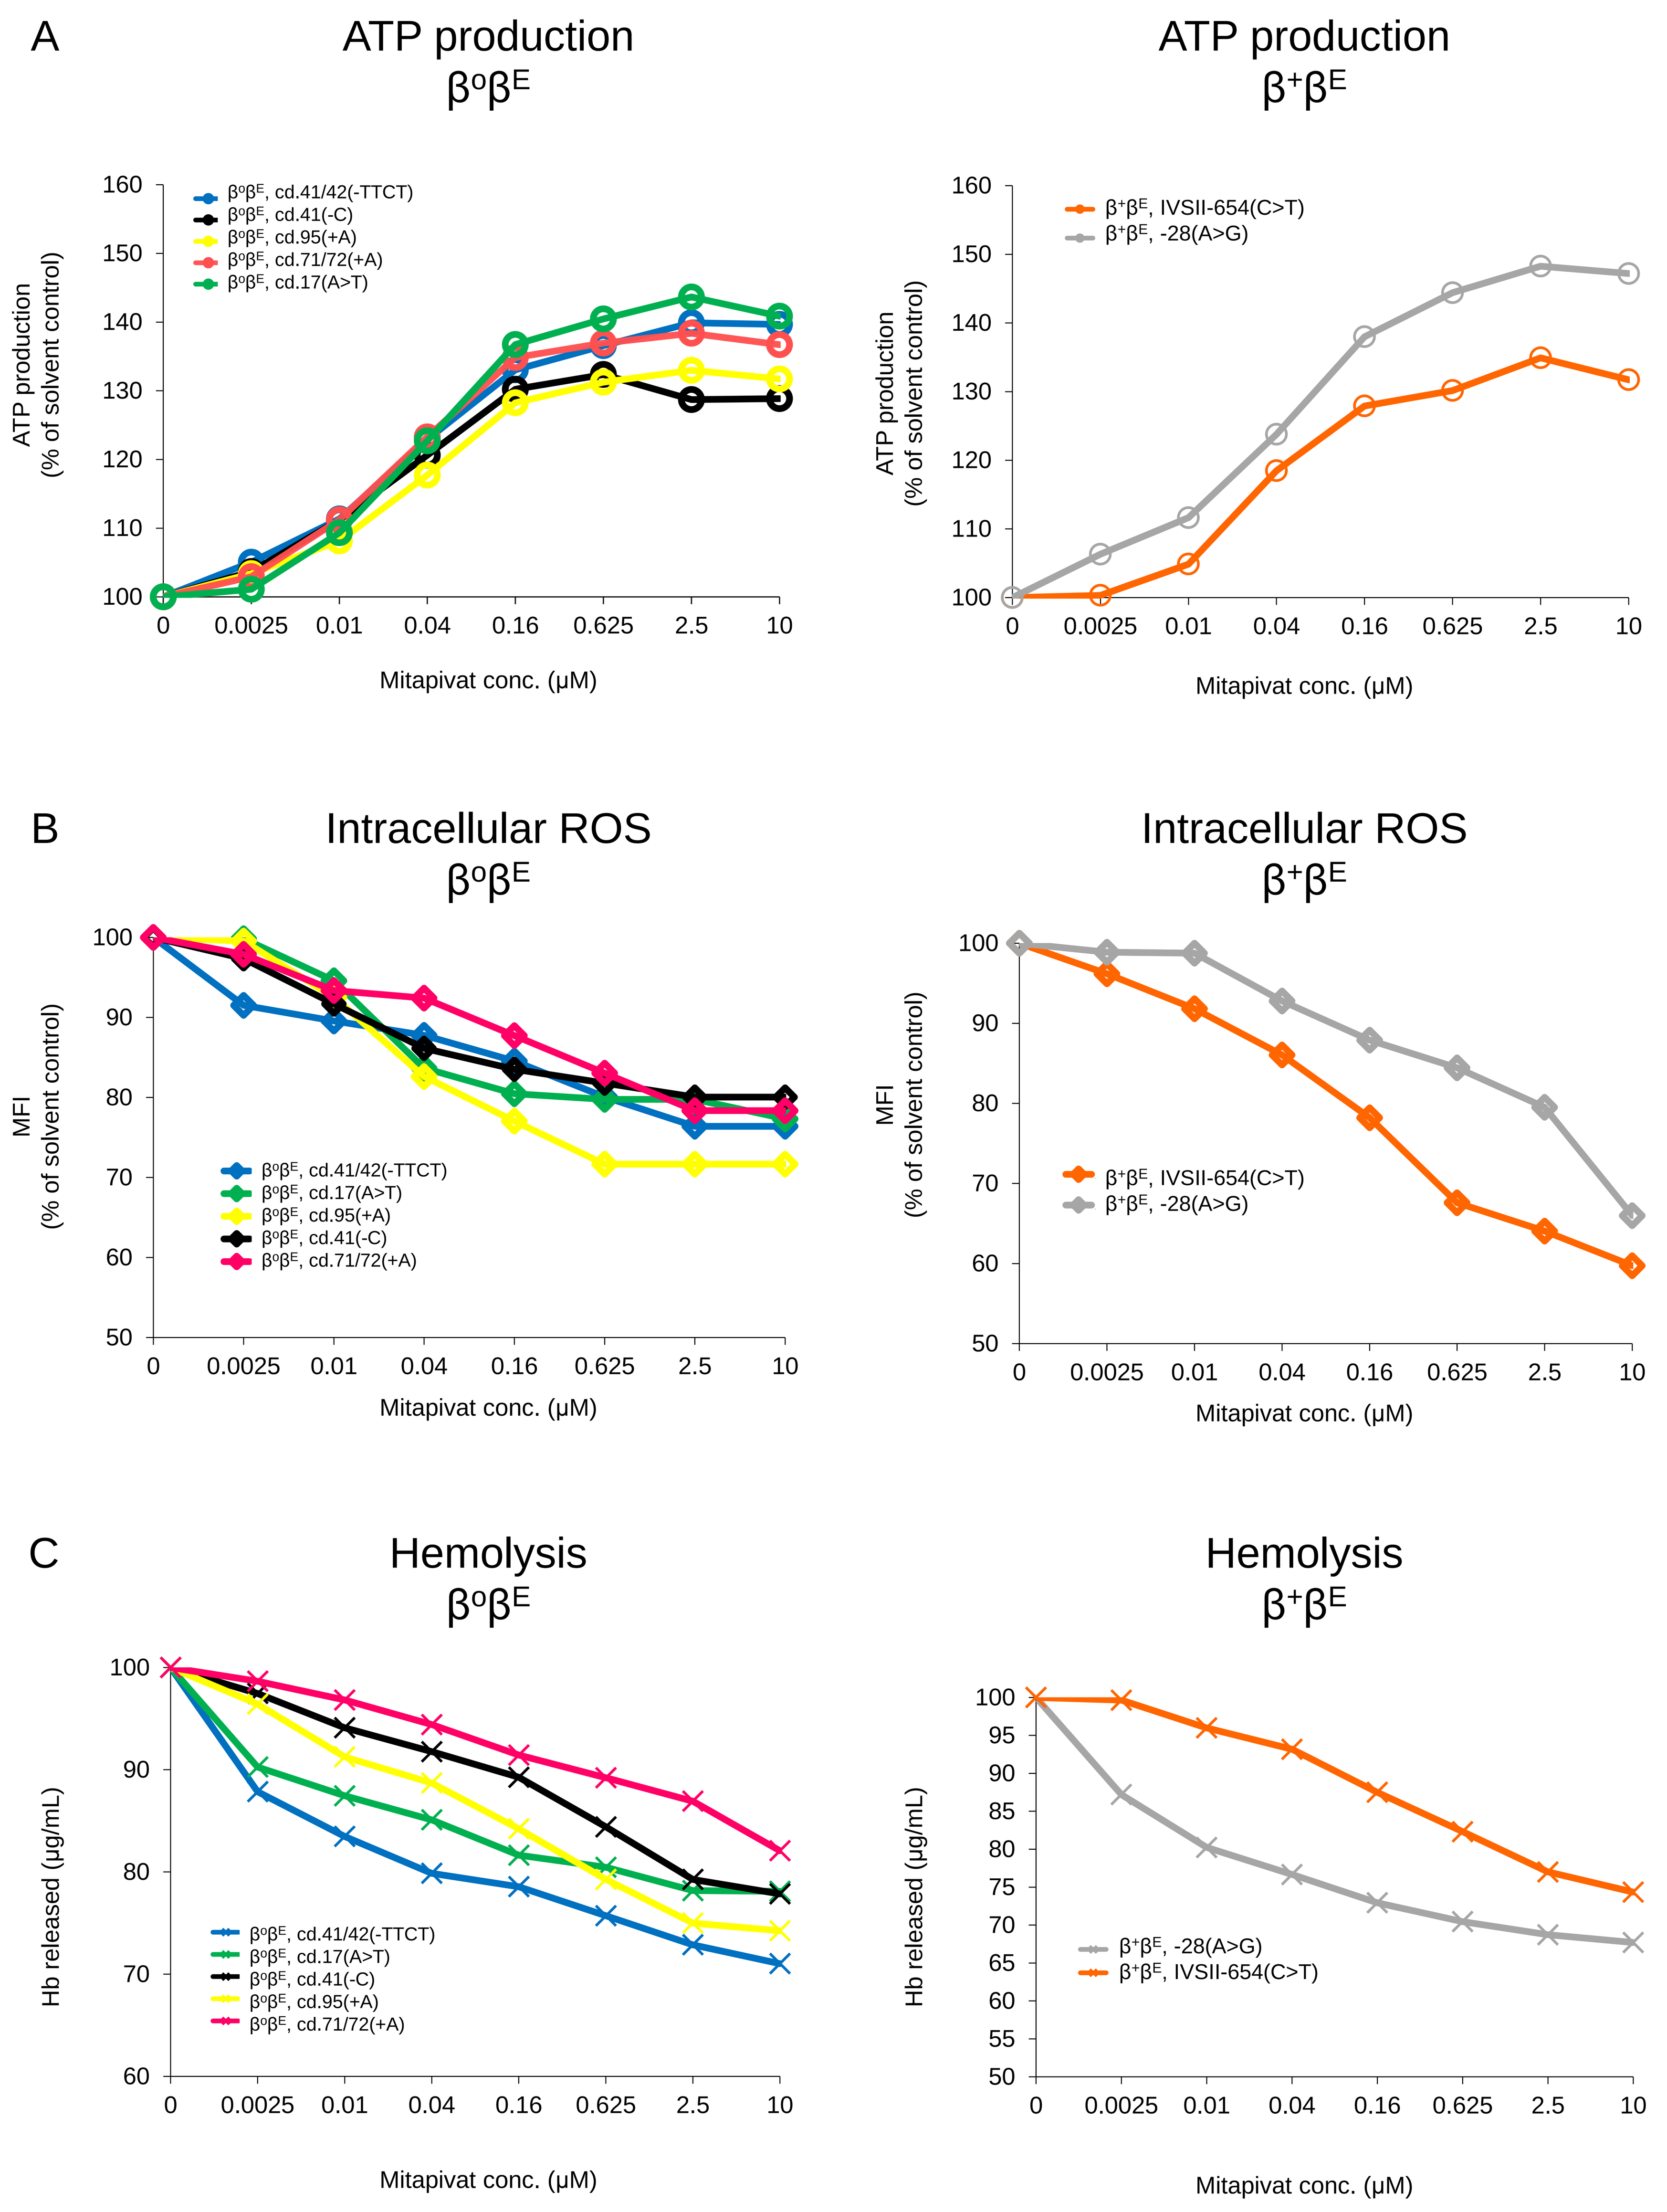

A
ATP production βoβE
ATP production β+βE
ATP production (% of solvent control)
βoβE, cd.41/42(-TTCT)
βoβE, cd.41(-C)
βoβE, cd.95(+A)
βoβE, cd.71/72(+A)
βoβE, cd.17(A>T)
β+βE, IVSII-654(C>T)
β+βE, -28(A>G)
Mitapivat conc. (µM)
B
Intracellular ROS βoβE
Intracellular ROS β+βE
MFI (% of solvent control)
C
Hemolysis βoβE
Hemolysis β+βE
Hb released (µg/mL)

Table 1 Baseline characteristics of 25 NTDT patients with β-thal/Hb E disease

Parameter	Normal range	β-globin genotype $\beta^o\beta^E$	$\beta^+\beta^E$	Interpretation
Sample size N (%)		20 (80%)	5 (20%)	
Age (years ± SD)		10.6 ± 6.6	11.6 ± 9.9	
Sex: N (%)		F: 14 (56%) M: 6 (24%)	F: 3 (12%) M: 2 (8%)	
CBC (mean ± SD)	**Normal range**			
• RBC (x10⁶/µL)	3.3 - 4.9	4.3 ± 0.6	5.0 ± 1.1	Normal
• Hb (g/dL)	11.5 - 15.5	8.0 ± 0.6	8.9 ± 1.0	↓↓
• Hct (%)	35.0 - 45.0	25.2 ± 1.8	27.7 ± 3.8	↓↓↓
• MCV (fl)	80.0 - 95.0	59.4 ± 6.3	56.3 ± 6.4	↓↓↓
• MCH (pg)	24.6 - 30.9	18.7 ± 2.2	18.0 ± 2.3	↓↓↓
• MCHC (g/dL)	31.0 - 37.0	31.7 ± 1.6	32.0 ± 0.9	↓
• RDW (%)	11.7 - 15.0	29.7 ± 2.7	23.3 ± 4.6	↑↑↑
•NRBC (/100 WBCs)	0	15.6 ± 51.4	0.5 ± 1.0	↑↑↑
•Ret (%)	0.5 - 2.0	3.6 ± 1.3	3.5 ± 1.3	↑↑
β-globin mutation N (%)		cd.41/42 (-TTCT): 12 (48%) cd.17 (A>T): 5 (20%) cd.71/72 (+A): 1 (4%) cd.95 (+A): 1 (4%) cd.41 (-C): 1 (4%)	IVSII-654 (C/T): 2 (8%) -28 (A>G): 3 (12%)	

Abbreviation: SD, standard deviation; F, female; M, male; CBC, complete blood count; RBC, red blood cell; Hb, hemoglobin; Hct, hematocrit; MCV, mean corpuscular volume; MCH, mean corpuscular hemoglobin; MCHC, mean corpuscular hemoglobin concentration; RDW, red cell distribution width; NRBC, nucleated red blood cell; WBC, white blood cell; Ret, reticulocyte; cd., codon

ACKNOWLEDGEMENTS

I would like to thank Agios Pharmaceuticals, Inc. for supplying the mitapivat compound to perform *ex vivo* experiments in this study.

CONTACT INFORMATION

Thidarat Suksangpleng, Ph.D.
Siriraj-Thalassemia Center, Faculty of Medicine Siriraj Hospital, Mahidol University, Bangkok, THAILAND, 10700
Email address: thidaratsuksangpleng@gmail.com, thidarat.suk@mahidol.ac.th