Dual activation of PKR and PKM2 reduced the development of fibrosis and iron deposition in a sickle cell disease nephropathy mouse model

P1107

Trang Nguyen, ScB[1], Christy Ingersoll, MBA[1], Kate Lizotte, ScB[2], Chi-Chao Chen, MD, PhD[2], Erin Tsai, MS[1], Megan Wind-Rotolo, PhD[1], Hui Gao, PhD[1]

[1]Agios Pharmaceuticals, Inc., Cambridge, MA, USA; [2]Employee of Agios Pharmaceuticals, Inc. at time of research

BACKGROUND

Sickle cell disease (SCD) and sickle cell nephropathy (SCN)

- SCD can lead to functional and structural kidney abnormalities that are collectively known as SCN[1–3]
- SCN is considered one of the most severe complications of SCD and is associated with a poor prognosis[1]
- SCN manifests in different forms, including albuminuria, deteriorating glomerular filtration rate, fibrosis, and progression to chronic kidney disease and kidney failure,[1,2] accounting for 16–18% of mortality in patients with SCD[3]
- In patients with SCN, improving anemia and reducing sickling and hemolysis through the activation of red blood cell-specific pyruvate kinase (PKR) may help lessen kidney damage by improving perfusion, reducing sickling-related ischemia, and minimizing the direct toxicity of free heme[4]
- Additionally, preclinical studies have shown that pyruvate kinase muscle isoenzyme 2 (PKM2) activation renders renal protective effects in diabetic kidney disease[5] and acute kidney injury[6]

Animal models of SCN

Berkeley sickle cell (BSC) disease mouse model[7]

- Repeated intravenous (IV) hemin challenges in SCD mice can cause acute hemolytic events and damage to endothelial cells, possibly leading to chronic kidney damage
 - Hemin-induced sickle cell acute kidney injury is reversible with a single challenge of hemin[8]

Experimental autoimmune glomerulonephritis (EAG)[9–11]

- EAG is induced by immunization with antigenic material from the glomerular basement membrane (GBM) (rat anti-GBM in mice)
- Renal pathology resembles human Goodpasture's disease, an autoimmune disorder characterized by rapidly progressive glomerulonephritis (with crescent formation) and glomerulosclerosis

Pyruvate kinase (PK) activators

Mitapivat

- Mitapivat is a first-in-class, oral, small-molecule allosteric activator of PK, including the PKR and PKM2 isoforms, that is under investigation for the treatment of SCD[4]
 - Mitapivat is approved in the US for the treatment of hemolytic anemia in adults with PK deficiency, and in the EU and the UK for the treatment of PK deficiency in adult patients[12–14]

Tebapivat (formerly AG-946)

- Tebapivat is an investigational, potent, oral activator of PK, including the PKR and PKM2 isoforms, that is under investigation in clinical trials for the treatment of SCD and lower-risk myelodysplastic syndrome[15]

OBJECTIVE

- **To investigate the therapeutic benefit of mitapivat and tebapivat, activators of PKR and PKM2 under investigation for the treatment of SCD, in preserving renal function in 2 mouse models of renal injury**

METHODS

Hemin-induced renal fibrosis in the BSC mouse model

- In this model, 11-week-old BSC mice were injected with either vehicle or hemin (7 µmol/kg body weight; 5 times on alternate days) and monitored for 25 weeks (**Figure 1; Supplemental methods** [QR code])
 - Histologic changes were described based on their distribution, severity, and morphologic character using a severity scale of 0–5; some findings were noted as present (P) or absent (0) when a severity score was not applicable
 - Mitapivat was investigated using the BSC model

Anti-GBM nephritis mouse model

- To evaluate potential therapeutic benefits of PK activation in reducing glomerular damage, the anti-GBM nephritis model was studied (**Figure 1; Supplemental methods** [QR code])
 - Tebapivat was investigated using the anti-GBM model

Figure 1. BSC and anti-GBM models

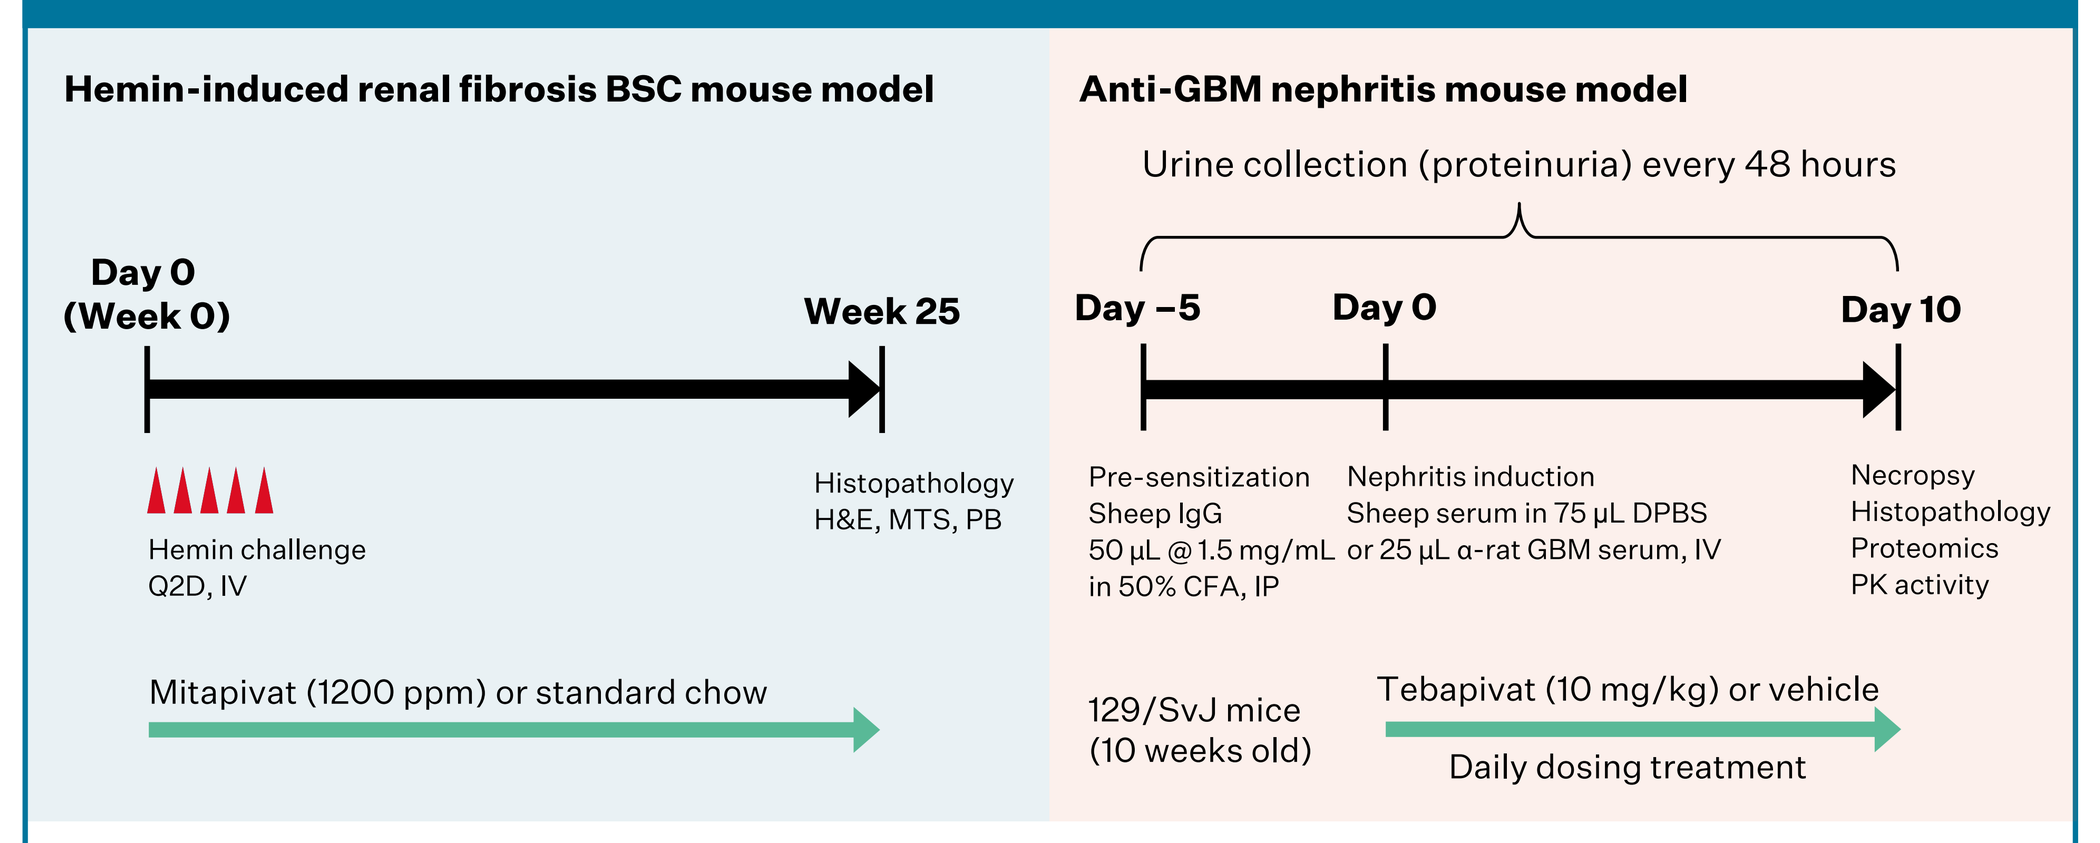
Hemin-induced renal fibrosis BSC mouse model
Day 0 (Week 0)
Week 25
Hemin challenge Q2D, IV
Histopathology H&E, MTS, PB
Mitapivat (1200 ppm) or standard chow
Anti-GBM nephritis mouse model
Urine collection (proteinuria) every 48 hours
Day –5
Day 0
Day 10
Pre-sensitization Sheep IgG 50 µL @ 1.5 mg/mL in 50% CFA, IP
Nephritis induction Sheep serum in 75 µL DPBS or 25 µL α-rat GBM serum, IV
Necropsy Histopathology Proteomics PK activity
129/SvJ mice (10 weeks old)
Tebapivat (10 mg/kg) or vehicle
Daily dosing treatment

BSC, Berkeley sickle cell; CFA, complete Freund's adjuvant; DPBS, Dulbecco's phosphate-buffered saline; GBM, glomerular basement membrane; H&E, hematoxylin and eosin; IgG, immunoglobulin G; IP, intraperitoneally; IV, intravenous; MTS, Masson's trichrome staining; PB, Prussian blue; PK, pyruvate kinase; Q2D, every 2 days

RESULTS

Hemin-induced renal fibrosis BSC mouse model

- Histologic analysis in the hemin-induced BSC mouse model showed that mice administered mitapivat had a numerically lower severity score for chronic progressive nephropathy compared with mice administered control chow and hemin (**Figure 2a**)
- Reduced protein casts were observed in the kidneys of mice treated with mitapivat compared with hemin controls (**Figure 2b**)

Figure 2. Incidence and severity of chronic progressive nephropathy (Figure 2a) and protein casts (Figure 2b)

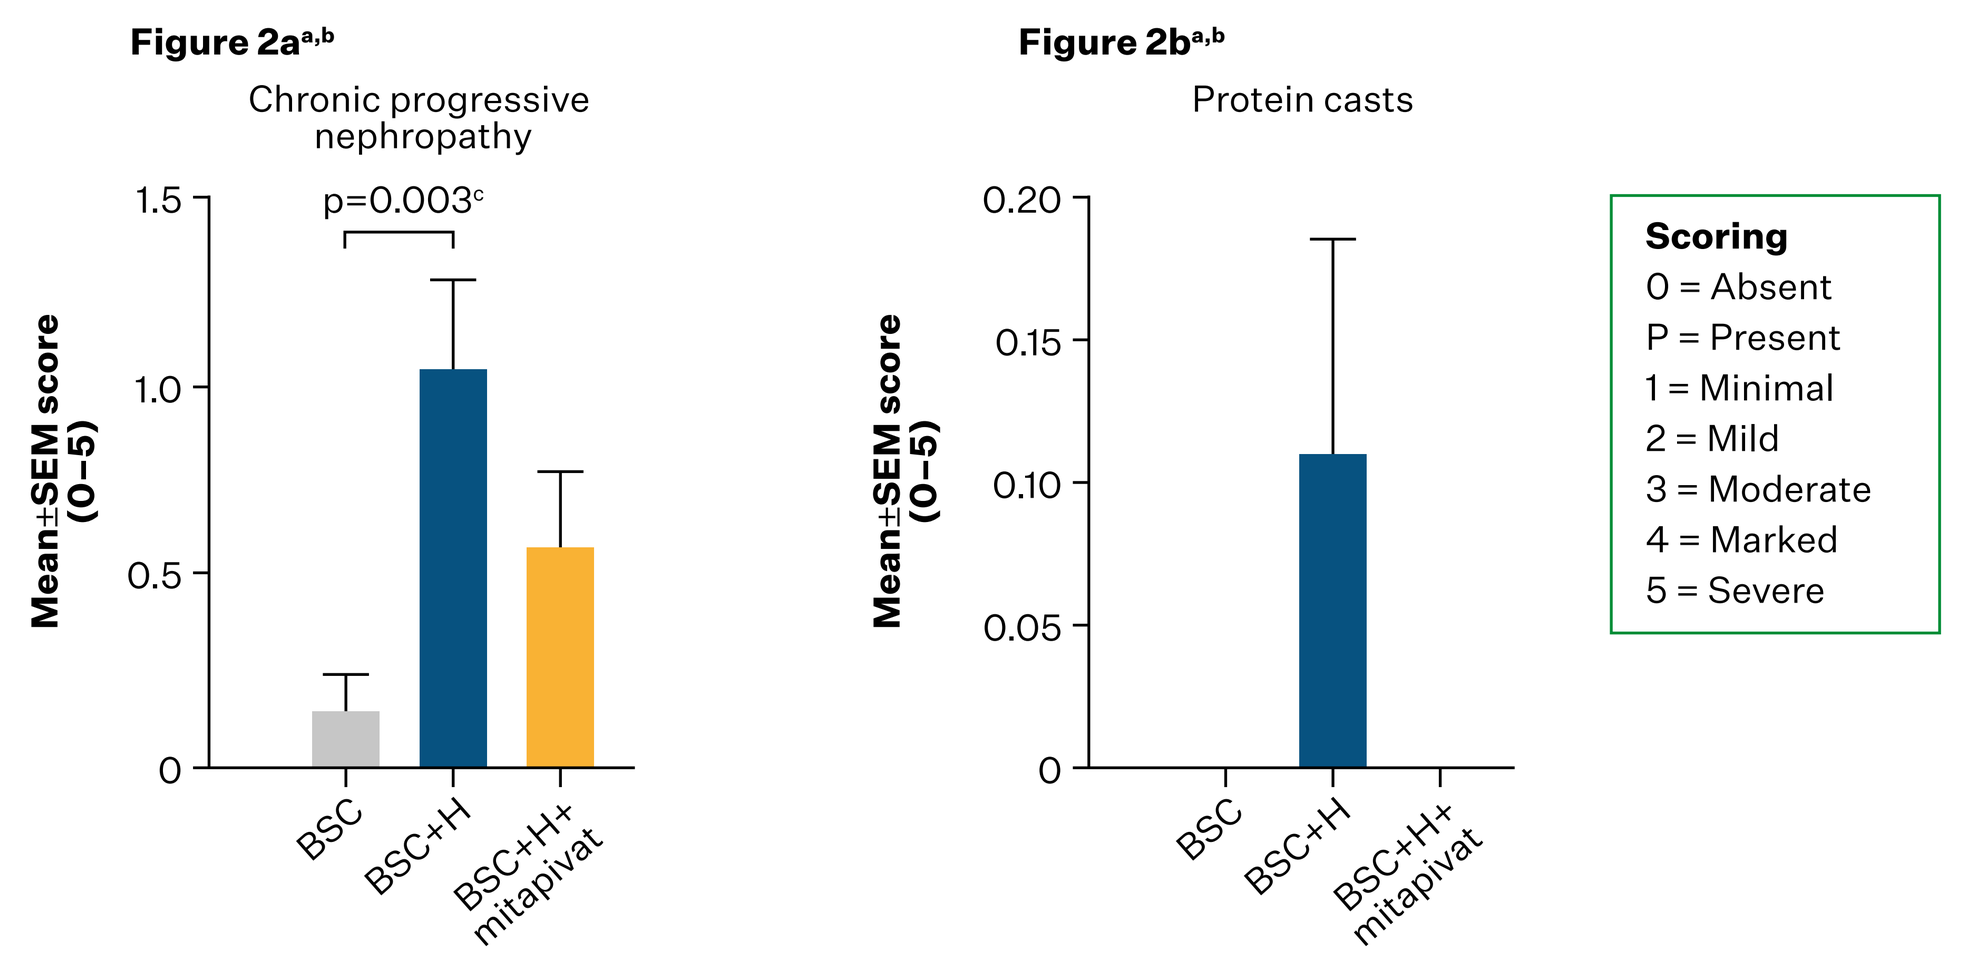
Figure 2a[a,b]
Chronic progressive nephropathy
p=0.003[c]
Mean±SEM score (0–5)
BSC
BSC+H
BSC+H+ mitapivat
Figure 2b[a,b]
Protein casts
Scoring
0 = Absent
P = Present
1 = Minimal
2 = Mild
3 = Moderate
4 = Marked
5 = Severe

[a]Evaluation of scoring was blinded; [b]n=12–18/BSC group (n=8 females; n=4–10 males); [c]One-way analysis of variance followed by Dunnett's multiple comparison test vs BSC+H
BSC, Berkeley sickle cell; H, hemin; SEM, standard error of the mean

RESULTS (CONT.)

- Histologic analysis in this model showed that the mitapivat-treated mice had a reduced occurrence and severity of iron deposition compared with standard chow-administered controls (**Figures 3 and 4**)
- The PB stain method is useful to detect the presence of non-hemin iron deposits
 - Ferric iron deposits stain bright blue, nuclei are red, and cytoplasm is pink
- BSC mice had a qualitatively greater amount of iron deposition in the cortical region of their kidneys compared with wild-type (WT) controls (**Figure 3**)
 - Administration of hemin did not affect iron deposition in the kidney cortex (**Figure 3**)
- Mitapivat-treated BSC mice showed reduced iron build-up and deposition in the kidney cortex of aged BSC mice (**Figure 3**)
- In a quantitative analysis, numeric decreases were observed in iron deposition in the kidney cortex and inner medulla in mitapivat-treated BSC mice compared with controls (**Figures 4a and b**)
- When female mice were excluded, mitapivat-treated BSC mice showed a significant reduction in iron deposition in the inner medulla compared with controls (staining score±SEM: 0.75±0.25 vs 1.70±0.21, p=0.014) (**Figure 4b**)

Figure 3. Presence of non-hemin iron deposits in the kidney using PB stain method

Iron deposition in the kidney cortex in 37-week-old BSC mice

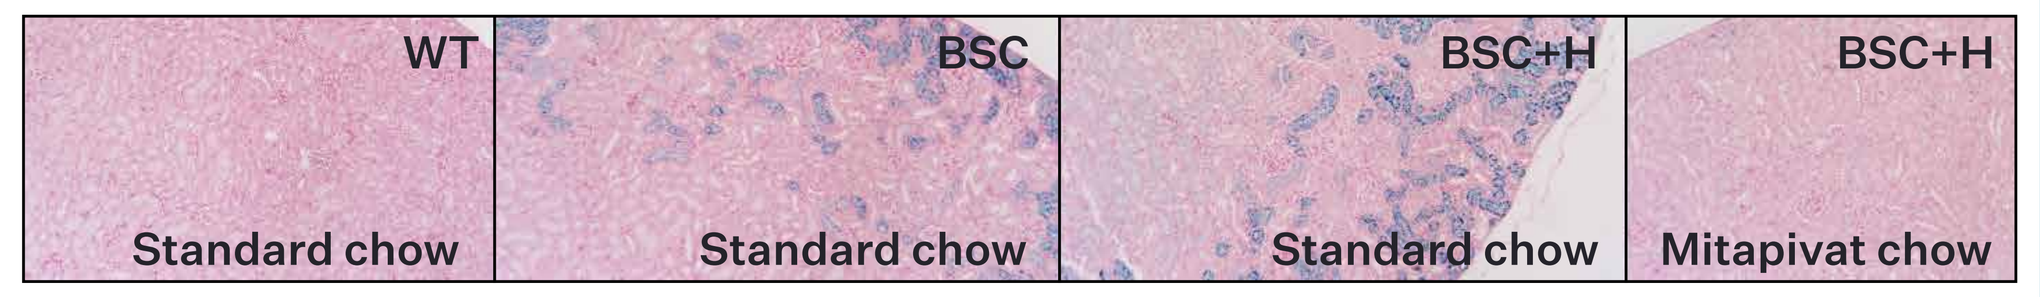
WT
Standard chow
BSC
Standard chow
BSC+H
Standard chow
BSC+H
Mitapivat chow

BSC, Berkeley sickle cell; H, hemin; PB, Prussian blue; WT, wild-type

Figure 4. Incidence and severity of iron deposition by region of the kidney of BSC mice (Figure 4a, 4b) and sex (Figure 4b)

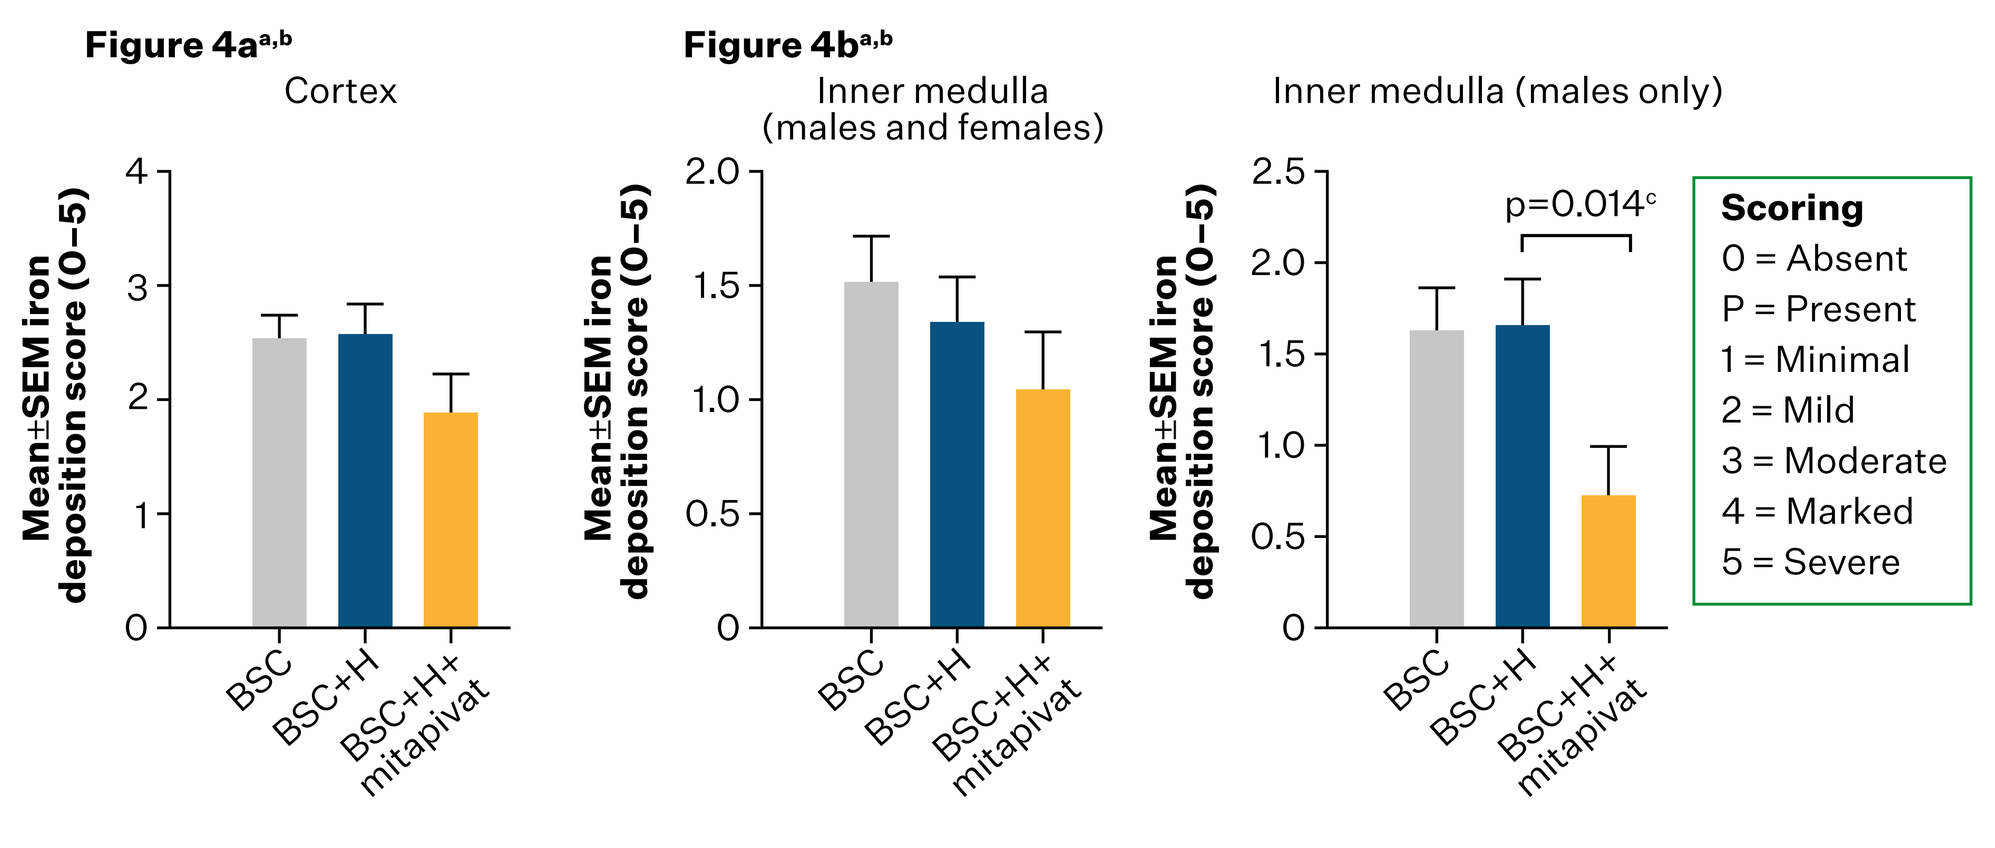
Figure 4a[a,b]
Cortex
Mean±SEM iron deposition score (0–5)
Figure 4b[a,b]
Inner medulla (males and females)
Inner medulla (males only)
p=0.014[c]
BSC
BSC+H
BSC+H+ mitapivat
Scoring
0 = Absent
P = Present
1 = Minimal
2 = Mild
3 = Moderate
4 = Marked
5 = Severe

[a]Evaluation of scoring was blinded; [b]n=12–18/BSC group (n=8 females; n=4–10 males);
[c]Mann–Whitney Test, BSC+H vs BSC+H+mitapivat
BSC, Berkeley sickle cell; H, hemin; SEM, standard error of the mean

- Histologic analysis in the hemin-induced BSC mouse model showed that the mitapivat-treated mice had a reduced severity of kidney fibrosis compared with chow-administered controls (**Figures 5 and 6**)
 - With MTS, collagen is stained blue, nuclei are dark brown, muscle is red, and cytoplasm is pink (**Figure 5**)
- Hemin aggravated interstitial fibrosis of the kidneys of 37-week-old BSC mice despite minimal presence of fibrosis, whereas mitapivat effectively reduced interstitial fibrosis (**Figure 5**)
- In a quantitative analysis, mitapivat-treated BSC mice had a significant reduction in fibrosis score compared with controls (0.17±0.17 vs 1.17±0.26, p≤0.002) (**Figure 6**)

Figures 5 and 6. Incidence and severity of kidney fibrosis

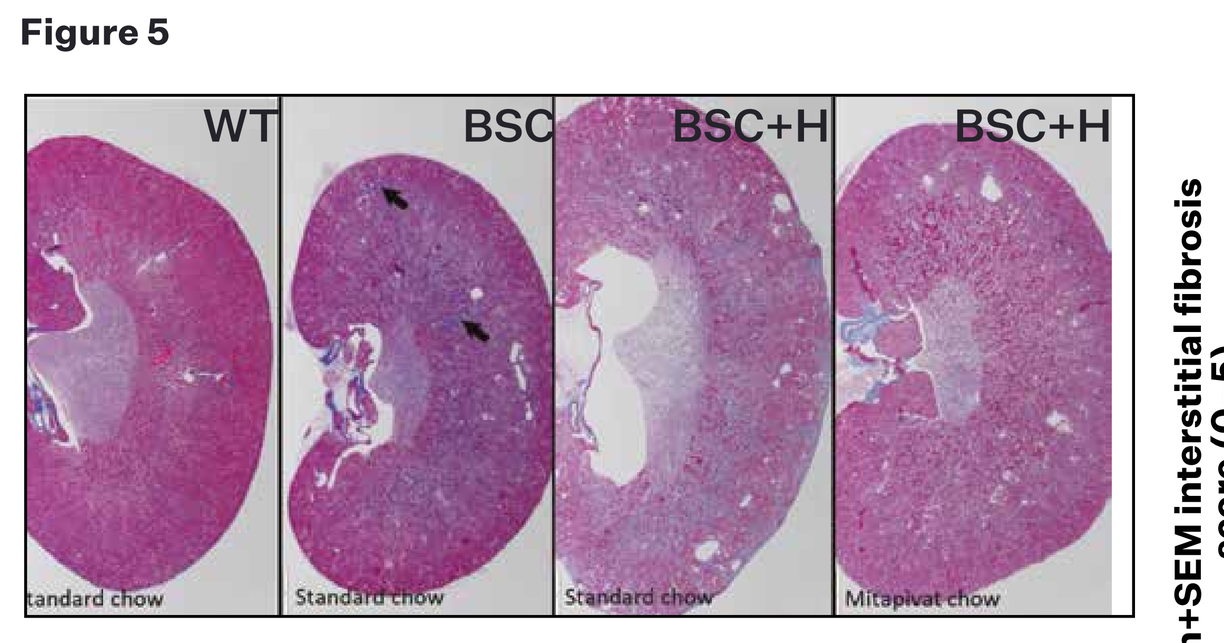
Figure 5
WT
BSC
BSC+H
BSC+H
Standard chow
Standard chow
Standard chow
Mitapivat chow

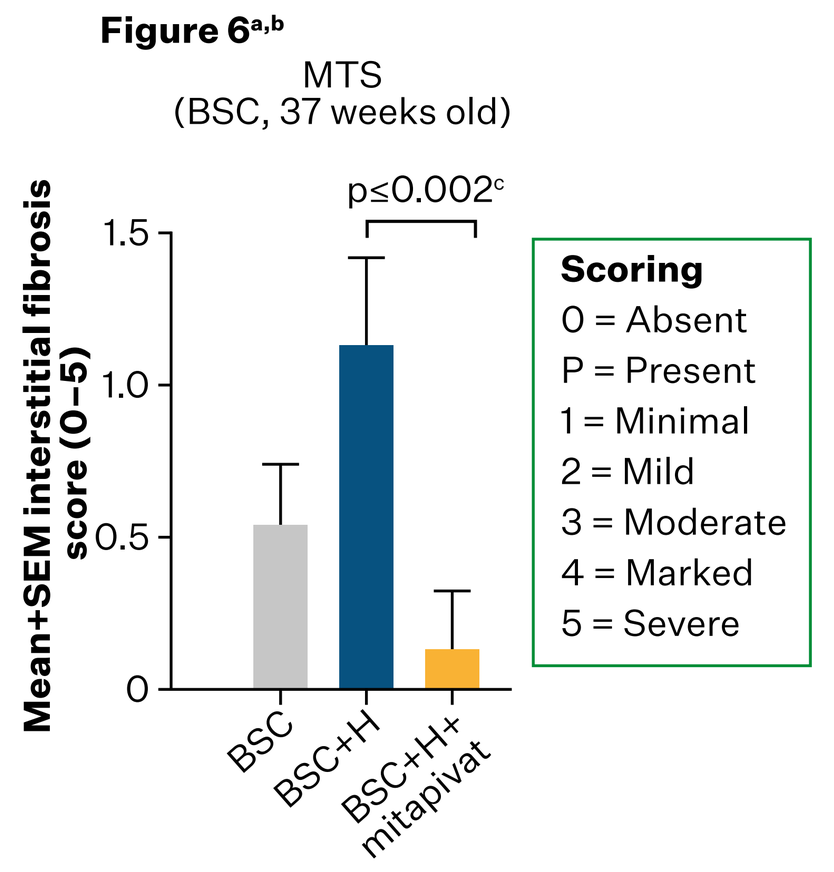
Figure 6[a,b]
MTS
(BSC, 37 weeks old)
p≤0.002[c]
Mean+SEM interstitial fibrosis score (0–5)
BSC
BSC+H
BSC+H+ mitapivat
Scoring
0 = Absent
P = Present
1 = Minimal
2 = Mild
3 = Moderate
4 = Marked
5 = Severe

[a]Evaluation of scoring was blinded; [b]n=12–18/BSC group (n=8 females; n=4–10 males); [c]Mann–Whitney Test, BSC+H vs BSC+H+mitapivat
BSC, Berkeley sickle cell; H, hemin; MTS, Masson's trichrome staining; SEM, standard error of the mean; WT, wild-type

Anti-GBM nephritis mouse model

PKM2 activity

- In the anti-GBM nephritis model, tebapivat treatment increased kidney PKM2 activity 5-fold compared with vehicle-treated mice (**Figure 7**)
- Tebapivat treatment significantly reduced the percentage of glomeruli with crescents compared with vehicle-treated mice (1.0%±0.3 vs 10.0%±2.4, p=0.004) (**Figure 8**)
 - No impact on proteinuria was observed, likely due to the short study duration of 10 days

Figures 7 and 8. Kidney PKM2 activity and proportion of glomeruli with crescents

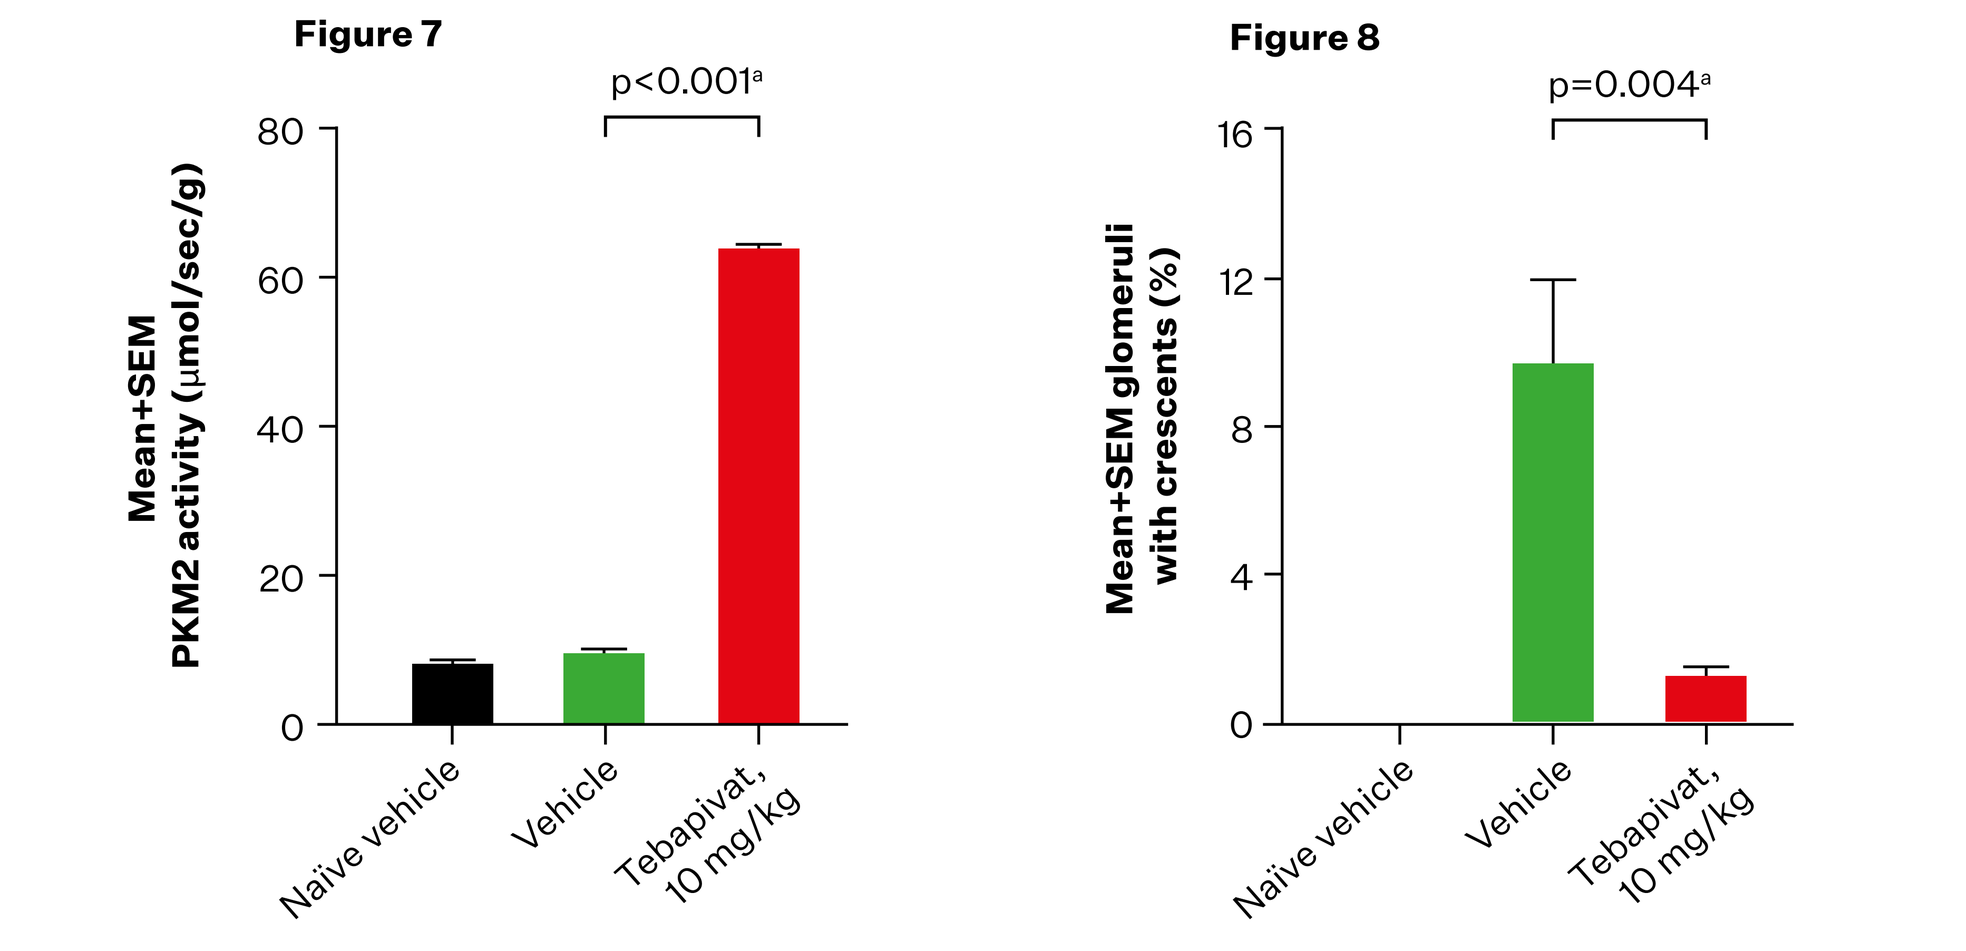
Figure 7
p<0.001[a]
Mean+SEM PKM2 activity (µmol/sec/g)
Naïve vehicle
Vehicle
Tebapivat, 10 mg/kg
Figure 8
p=0.004[a]
Mean+SEM glomeruli with crescents (%)
Naïve vehicle
Vehicle
Tebapivat, 10 mg/kg

[a]One-way analysis of variance followed by Dunnett's multiple comparison test vs vehicle
PKM2, pyruvate kinase muscle isoenzyme 2; SEM, standard error of the mean

- Proteomic evaluation of kidney tissue indicated that tebapivat downregulated several extracellular matrix proteins, including multiple collagens (Col1a1, Col1a2, Col3a1, Col5a2, Col6a6), elastin (Eln), and an integrin-binding protein (Fermt2) (denoted with a star in figure) compared with vehicle groups (**Figure 9**)
- Tebapivat treatment downregulated α-smooth muscle actin (Acta2) and myosin heavy chain (Myh11), markers for myofibroblast activation, suggesting that PK activation may modulate myofibroblast function in kidney disease

Figure 9. Proteomic analysis of tebapivat vs vehicle in an anti-GBM nephritis mouse model

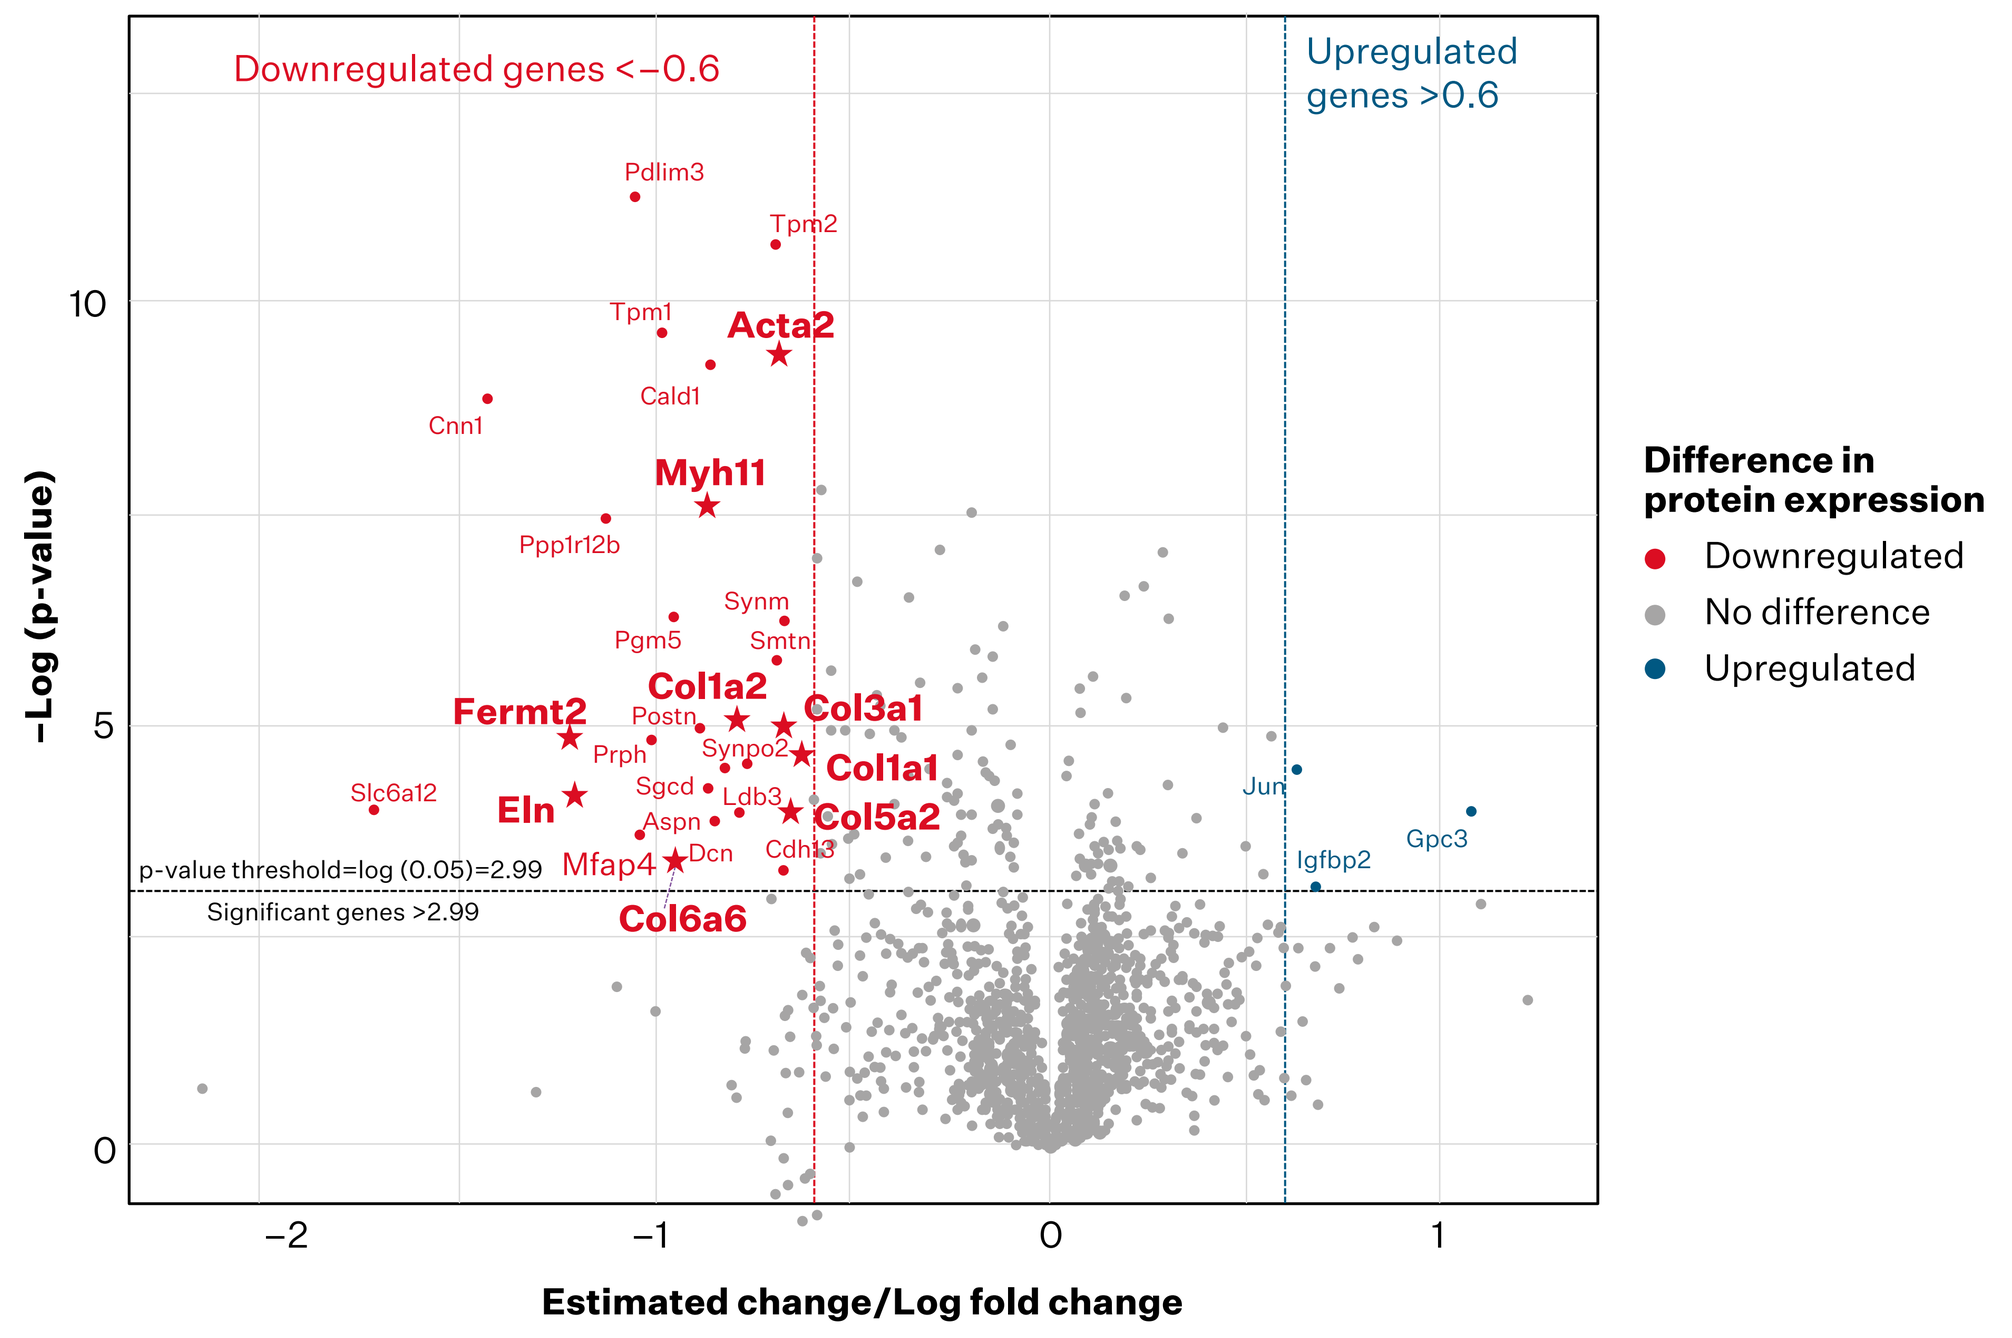
Downregulated genes <−0.6
Upregulated genes >0.6
Pdlim3
Tpm2
Tpm1
Acta2
Cald1
Cnn1
Myh11
Ppp1r12b
Synm
Pgm5
Smtn
Col1a2
Fermt2
Col3a1
Postn
Prph
Synpo2
Col1a1
Slc6a12
Eln
Sgcd
Ldb3
Col5a2
Aspn
Mfap4
Dcn
Cdh13
Col6a6
Jun
Igfbp2
Gpc3
p-value threshold=log (0.05)=2.99
Significant genes >2.99
−Log (p-value)
Estimated change/Log fold change
Difference in protein expression
Downregulated
No difference
Upregulated

GBM, glomerular basement membrane

CONCLUSIONS

- **PK activation by mitapivat significantly reduced the effects of hemin-induced chronic nephropathy and renal fibrosis, in addition to a reduced occurrence and severity of iron deposition, in an SCD mouse model, compared with controls**
- **PK activation by tebapivat significantly reduced crescent formation in the context of anti-GBM nephritis; tebapivat also upregulated PK activity in the kidney, with PKM2 activity increasing 5-fold, and downregulated several extracellular matrix proteins, compared with controls**
- **These findings underscore the potential of PK activators in improving kidney health by reducing hemolysis via PKR activation and slowing fibrosis development via PKM2 activation**
- **The potential role of PK activation in SCN is being further assessed in different clinical studies**

This study was funded by Agios Pharmaceuticals, Inc.

Acknowledgments: Medical writing assistance was provided by Julie Gray of Adelphi Group, Macclesfield, UK, funded by Agios Pharmaceuticals, Inc.

Referenes and supplemental material are available via the QR code

This study was funded by Agios Pharmaceuticals, Inc. Presented at the 66th American Society of Hematology (ASH) Annual Meeting and Exposition, December 7–10, 2024, San Diego, CA, USA, and virtual